PK Precision Estimation to Support the Design of a Pediatric Study of Belimumab Administered Subcutaneously

Richard Dimelow[1], Herbert Struemper[2]

[1]Clinical pharmacology Modelling and Simulation, GlaxoSmithKline, Gunnels Wood Road, Stevenage, SG1 2NY, UK
[2]Clinical pharmacology Modelling and Simulation, GlaxoSmithKline, Research Triangle Park, North Carolina, NC 27709, USA

Creation Date: 01/05/2019

Introduction

- Belimumab (BENLYSTA) is a monoclonal antibody that binds to the B-lymphocyte stimulator protein and is currently approved for the treatment of active systemic lupus erythematosus (SLE) in adults (IV and SC administration) and children (IV administration). To support the approval of belimumab administered subcutaneously for pediatrics, study 200908 is planned in a pediatric population with active SLE to investigate the safety, pharmacokinetics (PK) and pharmacodynamics (PD).

- Study 200908 (EudraCT number 2018-004645-16) will consist of two parts: Part A is a 12 week open-label treatment phase; Part B is an *optional* 40-week open-label continuation phase for any participant who completes Part A. To be conservative the sample size justification will be based on data from Part A only. The dose will be defined by body weight. This analysis specified that participants will receive 200 mg SC once a week (the adult approved dose) for ≥30 kg subjects, and once every two weeks for <30 kg subjects.*

- SLE is a rare disease in pediatrics and recruitment will be limited to 24 subjects. For ethical reasons blood samples for PK analysis will be kept to a minimum: 5 pre-dose blood samples (weeks 1, 2, 4, 8 and 12) followed by an 8 week washout sample (Part A of study 200908).

- To report the clearance (CL) and volume of distribution (V) it is a regulatory requirement for the 95% confidence interval (CI) to be within 60% and 140% of the median estimate for this interval with at least 80% likelihood [1]. Meeting this condition will provide confidence the study is sufficiently powered to estimate the PK in pediatrics, and to address the robustness against dropout. Based on the prior pediatric IV study for belimumab a worst case 25% drop-out rate was assessed.

Methods

- A **simulation / re-estimation** approach was used to assess PK parameter precision [1].

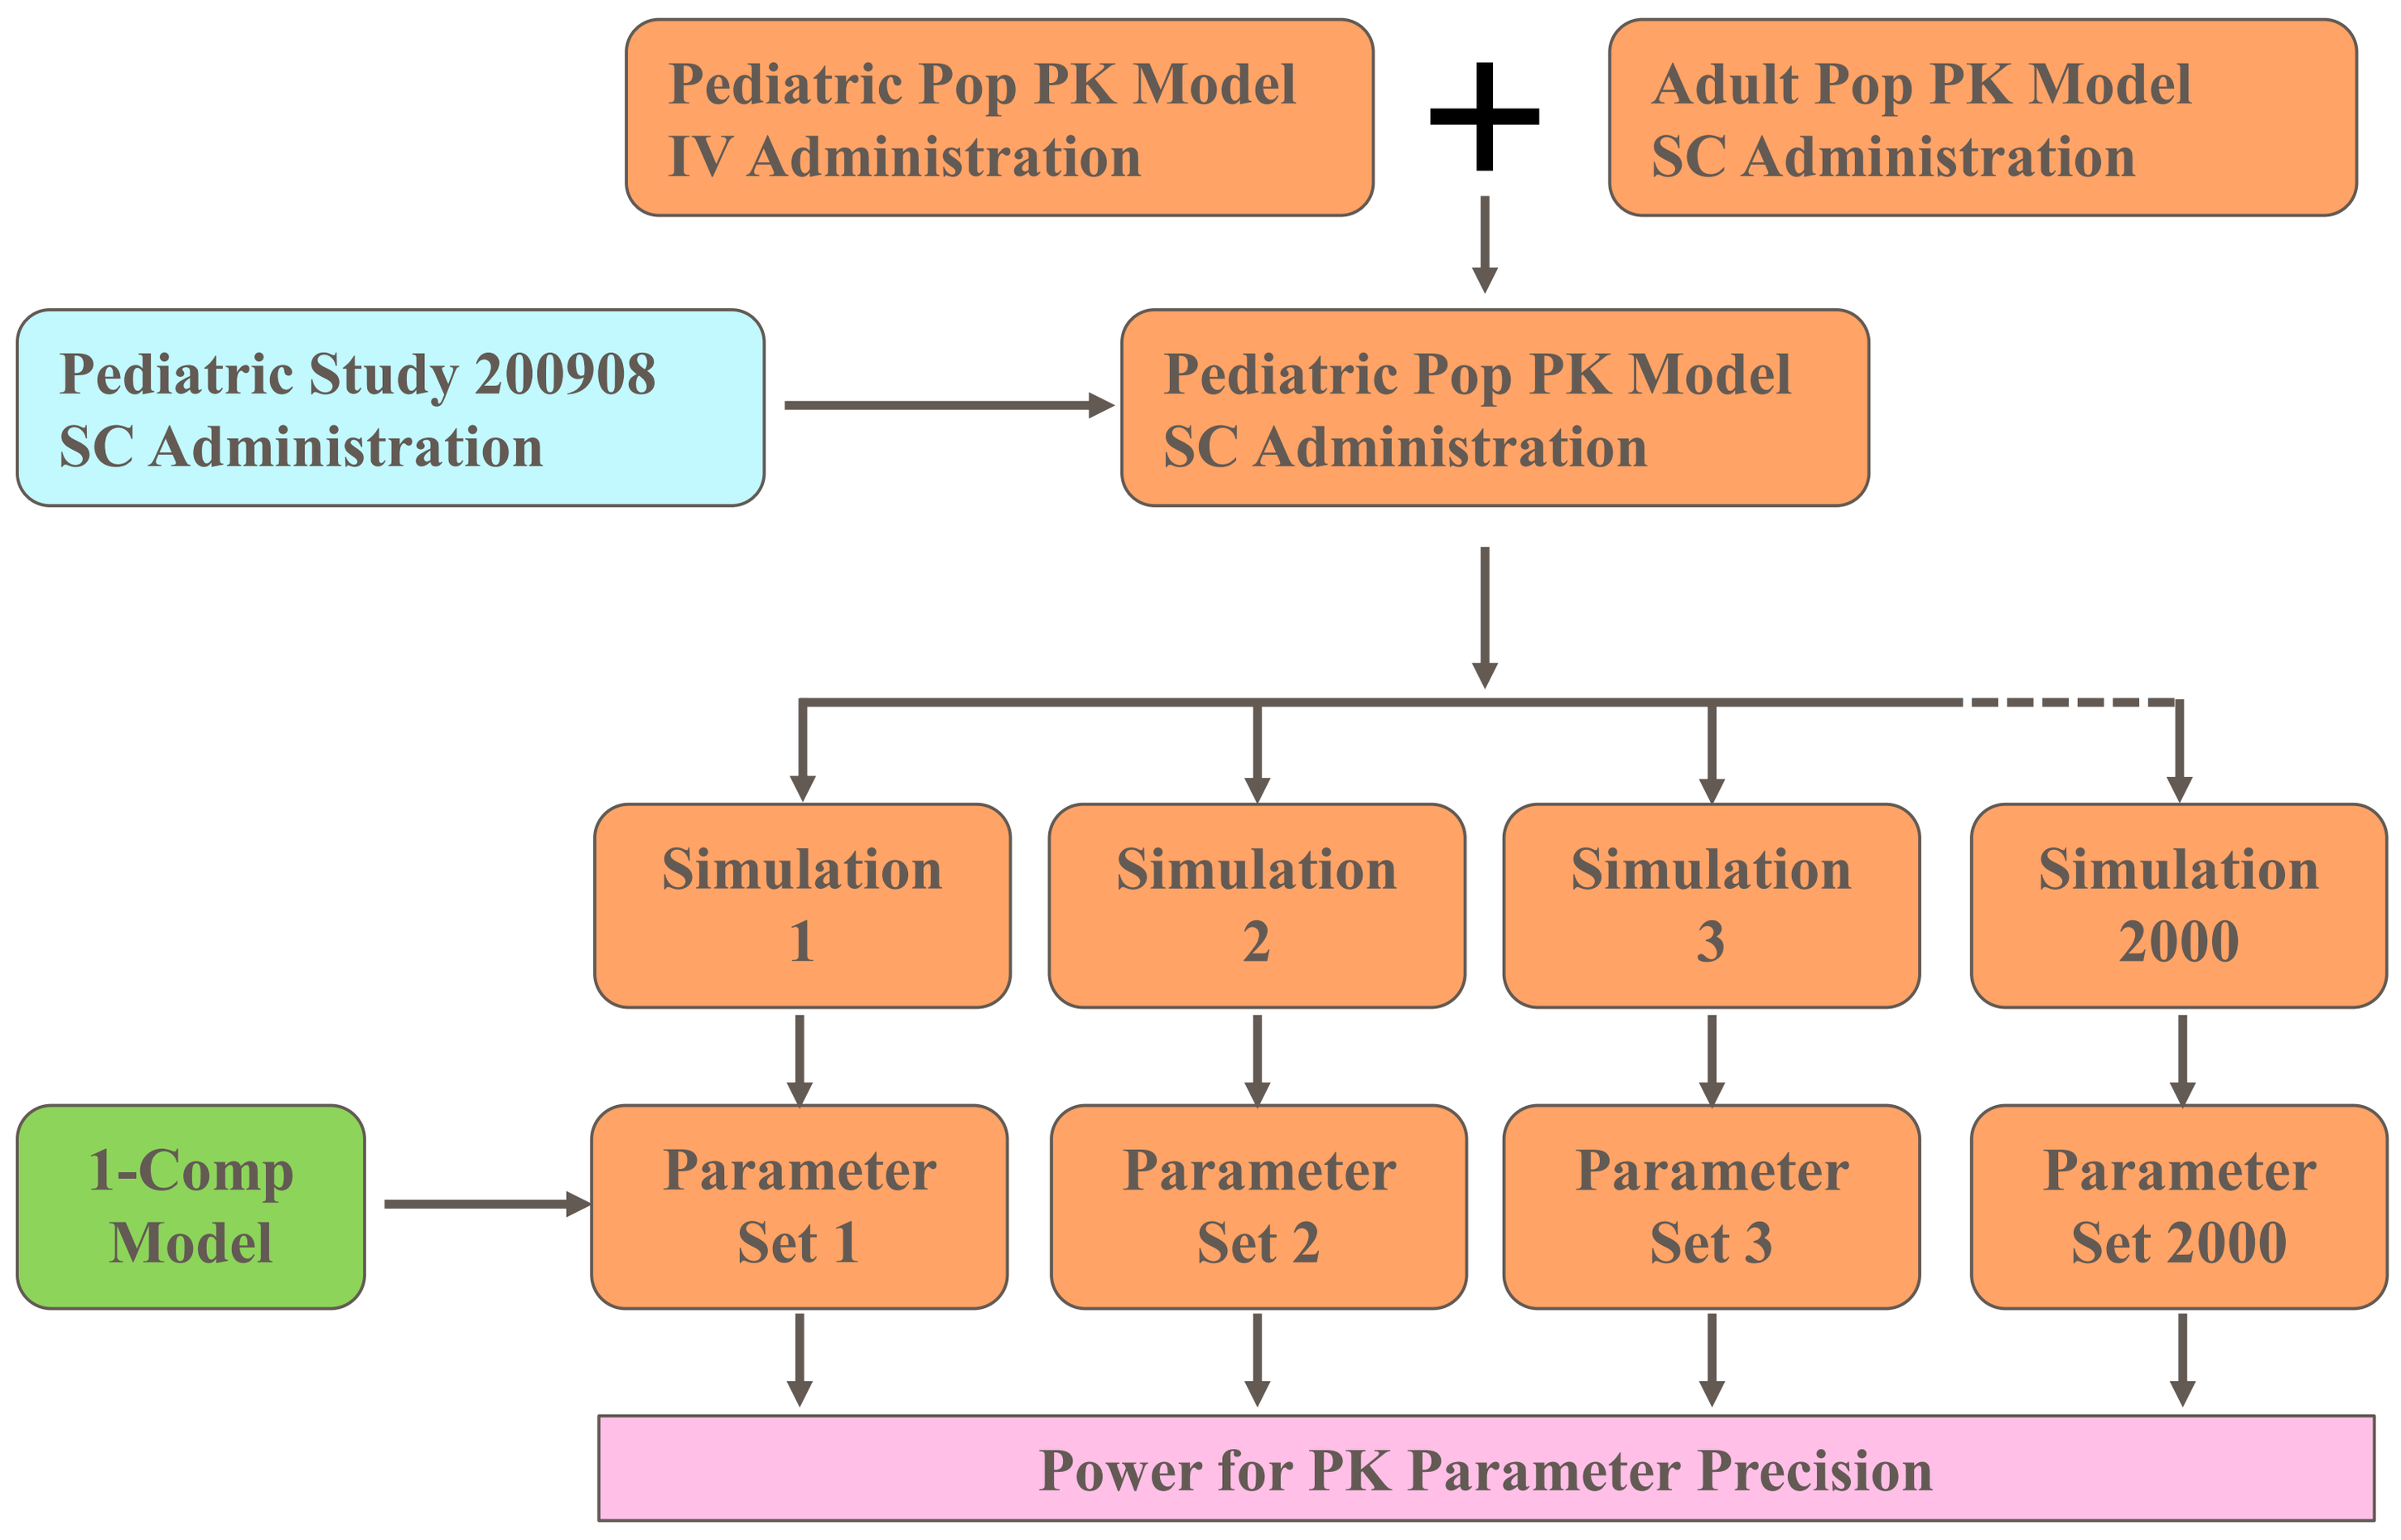
Pediatric Pop PK Model
IV Administration
+
Adult Pop PK Model
SC Administration
Pediatric Study 200908
SC Administration
Pediatric Pop PK Model
SC Administration
Simulation 1
Simulation 2
Simulation 3
Simulation 2000
1-Comp Model
Parameter Set 1
Parameter Set 2
Parameter Set 3
Parameter Set 2000
Power for PK Parameter Precision

- A pediatric population PK (popPK) model with SC administration was constructed, by combining a pediatric popPK model previously developed for IV administration (Study BEL114055) [2] with the absorption component of an adult SC popPK model for belimumab [3]. 2000 trial outcomes were simulated for the planned study 200908 (Part A only).

- A one-compartment PK model with first order absorption and elimination was fitted to each simulated trial outcome. Baseline fat-free mass (FFM) was previously shown to be the most pharmacokinetically relevant covariate in pediatrics [2] and included as a body-size covariate of clearance (CL) and volume of distribution (V).

Parameter	Parameterisation
CL or V Fixed Exponent Ka	$x = \theta_1 \times \left(\frac{FFM}{34.4}\right)^{0.75,\,1.0}$ *or* θ_1 with $cov(\theta_1) = \sigma_1^2$ $\frac{se(x)}{x} = \frac{\sigma_1}{\theta_1}$ $CI_{95} = \left(1 + t_{2.5} \times \frac{se(x)}{x},\ 1 + t_{97.5} \times \frac{se(x)}{x}\right)$
CL or V Estimated Exponent	$log(x) = \theta_1 + \theta_2 \times \left(\frac{FFM}{34.4}\right)$ with $cov(\theta_1, \theta_2) = \begin{pmatrix} \sigma_1^2 & \sigma_{12} \\ \sigma_{12} & \sigma_2^2 \end{pmatrix}$ $se(log(x)) = \sqrt{\sigma_1^2 + \sigma_2^2 \times \left(log\left(\frac{FFM}{34.4}\right)\right)^2 + 2 \times \sigma_{12} \times log\left(\frac{FFM}{34.4}\right)}$ $CI_{95} = \left(e^{t_{2.5} \times se(\log(x))},\ e^{t_{97.5} \times se(\log(x))}\right)$

- Each model fit provided a set of PK parameter estimates (θ) with covariance matrix for precision (cov(θ)), from which the 95% CI relative to the median estimate was derived (see table). Where precision depends on the FFM the CI was calculated at the age extremes (5 years and 17 years), and also at the typical value (FFM=34.4 kg), and the largest of these CI selected to represent the precision.

- $Power|_{60,140}$ was defined by the proportion of simulated results with 95% CI within 60% to 140%.

- $Power|_{80,120}$ was defined by the proportion of simulated results with 95% CI within 80% to 120%, and was a more stringent measure of precision to differentiate cases where $Power|_{60,140}$ is close to 100%.

- The simulation-based power calculations were compared with a corresponding theoretical value where PK sampling is sufficiently dense to accurately estimate individual CL and V parameters via non-compartmental analysis (NCA) [1].

***Based on the proposed dosing regimen at the time of the analysis. The final regimen is under review and subject to change.**

Results

		Power CI95 = 60% to 140%	Power CI95 = 80% to 120%
Model 1	FFM exponent on CL fixed at 0.75 FFM exponent on V fixed at 1.0 Ka fixed at 0.235 1/day	CL: 100% V: 100% Ka: 100%	CL: 95.2% V: 94.8% Ka: 100%
Model 2	FFM exponent on CL estimated FFM exponent on V estimated Ka estimated	CL: 35.9% V: 22.2% Ka: 8.5%	CL: 0.6% V: 0.2% Ka: 0%
Model 3	FFM exponent on CL estimated: prior = 0.75±0.1 FFM exponent on V estimated: prior = 1.0 ±0.1 Ka estimated: prior = 0.3 ±0.1 1/day	CL: 100% V: 99.6% Ka: 68.5%	CL: 57.6% V: 34.0% Ka: 5.1%

The effect of subject number on precision: Simulation / re-estimation using MODEL 3 (broken line) vs theoretical NCA analysis (solid line) power calculations for CI95 = 60% to 140% (red) and 80% to 120% (blue). Power = 80% threshold (horizontal, black, broken line).

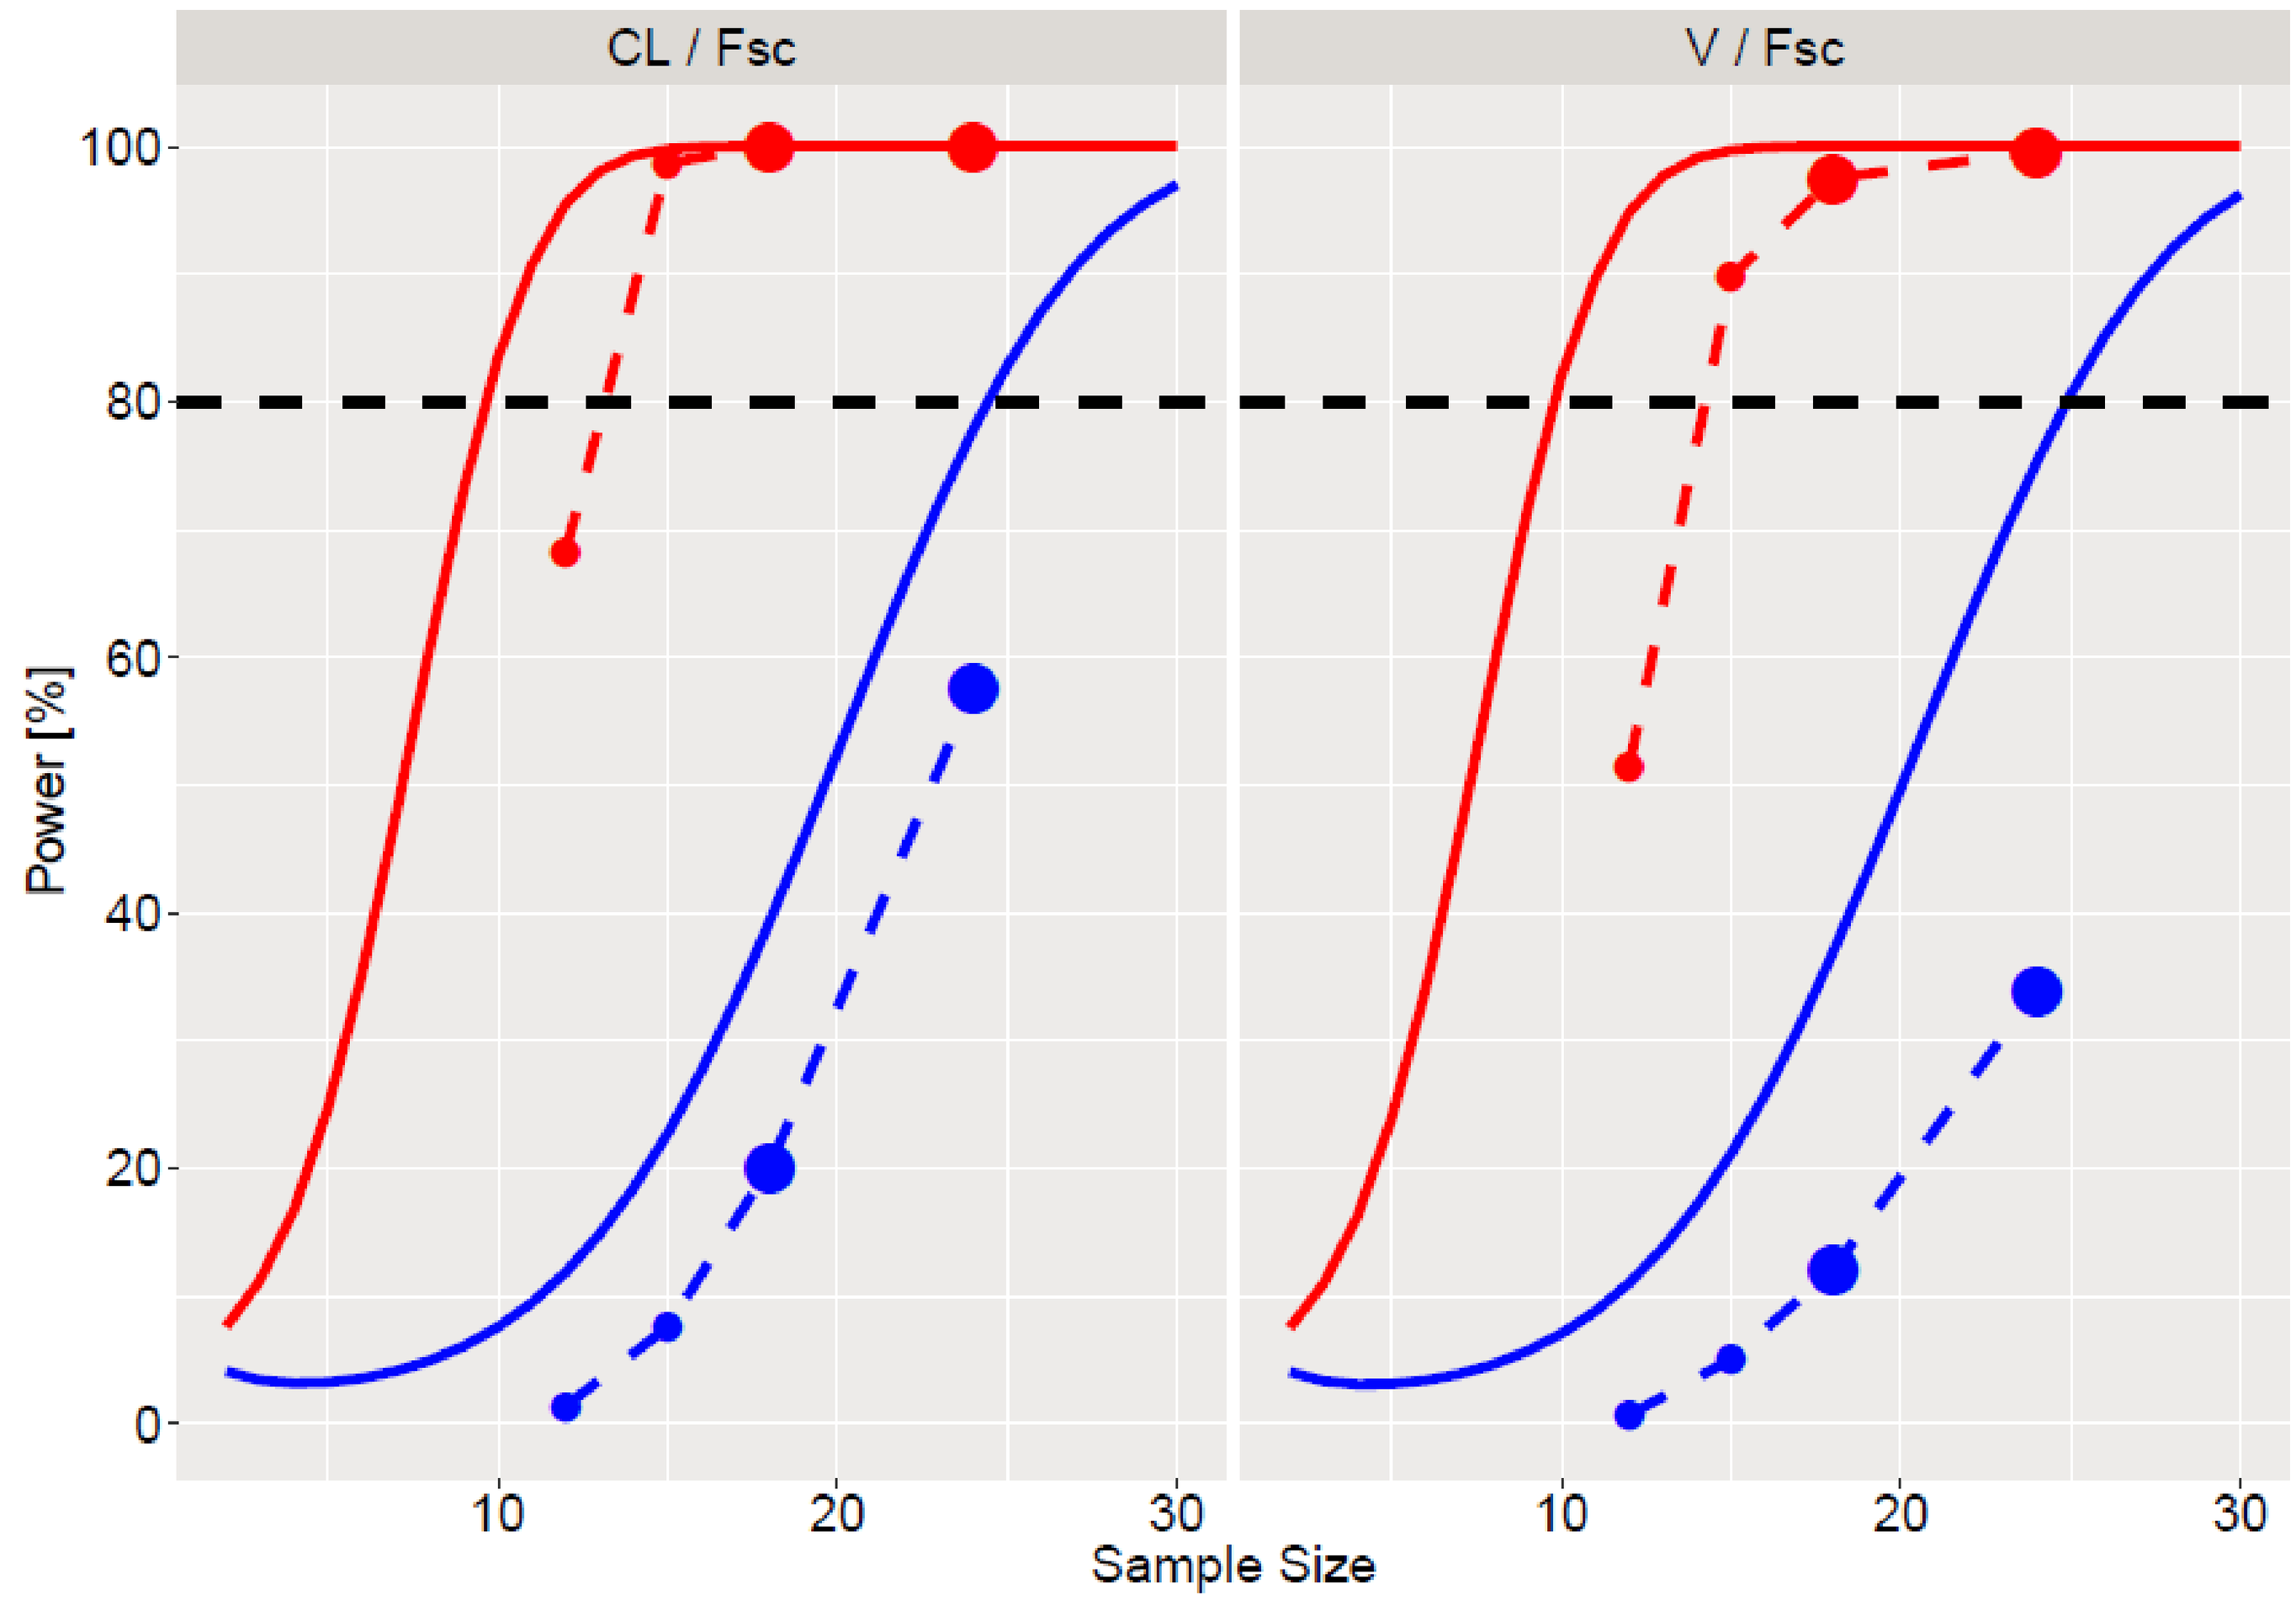
CL / Fsc
V / Fsc
Power [%]
0
20
40
60
80
100
10
20
30
Sample Size

- **MODEL 1**
- CL and V can be estimated with 100% power (CI95 = 60% to 140%) providing the FFM exponents on CL and V are fixed at their theoretical values (0.75 and 1), which are close to the values estimated from study BEL114055 (0.691 and 0.944 [2]). Even when measured against a higher level of precision (CI95 = 80% to 120%) the power remained above 90%.

- **MODEL 2**
- Estimating the FFM exponent on both CL and V had a negative impact on precision (CI95 = 60% to 140%) reducing the power to 35.9% (CL) and 22.2% (V).
- The power to estimate the absorption rate constant with CI95 = 60% to 140% was also small (8.5%), consistent with there being no PK samples planned around maximum concentration.

- **MODEL 3**
- Prior likelihoods on CL (0.75±0.1) and V (1.0±0.1) exponents and Ka (0.3±0.1 1/day) contained the model estimated values (0.691, 0.944 [2] and 0.235 1/day [3]) within 1 standard deviation.
- The prior likelihoods had a beneficial impact on precision (CI95 = 60% to 140%) increasing the power to 100% (CL) and 99.6% (V).
- Including a prior likelihood on Ka also improved the precision in its estimate, increasing the power (CI95 = 60% to 140%) from 8.5% (MODEL 2, no prior) to 68.5%, although the power is still lower than the 80% threshold required for reporting this parameter.
- The study design can accommodate substantial drop-out (from 24 to 15 subjects) and still be sufficiently powered (>80%) to estimate CL and V within CI95 = 60% to 140%.

- **Non-Compartmental Analysis**
- The alternative rich PK sampling scheme which enables accurate individual CL and V estimates via NCA, suggests N=10 is the minimum sample size for the study to be powered to estimate CL and V.

CONCLUSIONS

- The presented simulation/re-estimation study confirmed that with the PIP/PSP agreed sample size (N=24) Study 200908, designed with sparse PK sampling to minimize patient burden, is sufficiently powered to estimate CL and V with the required level of precision.
- Power results depended strongly on the parameterization of the PK model selected for re-estimation.
- Model options with body size exponents fixed to theoretical values, or informed by a prior likelihood (0.75±0.1 for CL and 1.0±0.1 for V, justified in this case by [2]), resulted in sufficient power for the assessed trial design.
- The study is robust against drop-out, and would be powered to estimate CL and V for N≥15. There is minimal loss of power for N=18, which corresponds to a 25% drop-out rate.
- For a rich sampling scheme where individual PK parameters can be accurately measured via NCA, a minimum of 10 subjects would be required to estimate CL and V with 80% power. This is a best case scenario that would typically not be achievable for long-half-life drugs in pediatric clinical practice, where patient burden related to sampling needs to be minimized.

References

[1] Wang Y, Jadhav P R, Lala M and Gobburu J V. Clarification on Precision Criteria to Derive Sample Size When Designing Pediatric Pharmacokinetic Studies. The Journal of Clinical Pharmacology 52: 1601. 2012. DOI: 10.1177/0091270011422812
[2] Struemper H, Dimelow R. Population Pharmacokinetics of Belimumab Administered Intravenously in Children with Systemic Lupus Erythematosus. PAGE abstract 8985, 2019
[3] Struemper H, Thapar M, Roth D. Population Pharmacokinetic and Pharmacodynamic Analysis of Belimumab Administered Subcutaneously in Healthy Volunteers and Patients with Systemic Lupus Erythematosus. Clin Pharmacokinet. 57(6):717-728, 2018. DOI: 10.1007/s40262-017-0586-5.

Study 200908 is sponsored by GlaxoSmithKline (EudraCT number 2018-004645-16)
R Dimelow and H Struemper are employees and share holders of GlaxoSmithKline

Created by: Digital Media Services